Optimization and Characterization of ATA3219: A Novel and Potent Allogeneic CD19-CAR T Therapy Without Gene Editing

Christina Pham[1], Alfonso Brito[1], Morgan Charbonneau[1], Ania Habibi[1], Mackenzie Sober[1], Cheng-Jang Wu[1], Cokey Nguyen[1]

[1]Atara Biotherapeutics, Inc., Thousand Oaks, CA

Poster 237

BACKGROUND

- EBV T cells represent a unique approach for generating off-the-shelf therapies and are the first approved allogeneic T cell immunotherapy with the EU approval of tabelecleucel. Unlike gene-edited approaches aimed to inactivate TCR function to reduce the risk for GvHD, EBV T cells maintain expression of native T-cell receptors that promote *in vivo* persistence[1], while also demonstrating inherently low alloreactivity due to their recognition of defined viral antigens.
- Allogeneic CD19-directed CAR EBV T cells have previously demonstrated encouraging efficacy and a favorable safety profile with no supportive evidence for GvHD or high-grade cytokine release syndrome.[2]
- ATA3219 is a next-generation allogeneic CD19 targeted CAR containing the novel 1XX CD3ζ signaling domain, built on Atara's EBV T-cell platform. The 1XX domain contains mutations of 2 tyrosine residues to 2 phenylalanines in immunoreceptor tyrosine-based activation motif (ITAM) 2 and ITAM3, impeding their phosphorylation and downstream signaling. This 1XX domain is designed to extend CAR T-cell persistence by sustaining T-cell effector functions without eliminating or compromising their potency[3], and has been associated with favorable response rates, safety and durability.[4]
- We have optimized ATA3219 from a previously reported process[5] to enrich for a less differentiated phenotype clinically correlated to durability.[6] Process improvements prioritizing a predominant memory immunophenotype yielded product lots that showed robust expansion and potent anti-tumor efficacy in preclinical models.

METHODS

- We generated ATA3219, a CD19-targeted CAR containing a modified CD3ζ signaling domain, 1XX, built on our EBV T-cell platform without modification of the endogenous TCR. To serve as a benchmark comparator, we additionally produced autologous CD19 CAR T cells using a clinically relevant 12-day process from CD3/CD28-activated T cells. Functional assessments were conducted to characterize the phenotype, *in vitro* potency, proliferation, cytokine response and *in vivo* anti-tumor efficacy.

Generation of EBV T cells expressing CD19-1XX CAR

Figure 1. PBMCs are isolated following healthy donor leukapheresis. The B-cell fraction is transformed with EBV, generating an EBV+ lymphoblastoid cell line (BLCL). PBMCs are stimulated with BLCLs prior to retroviral introduction of the CD19-targeted CAR with a 1XX signaling domain. CD19-1XX CAR+ EBV T cells (ATA3219) are expanded and then harvested for downstream use.

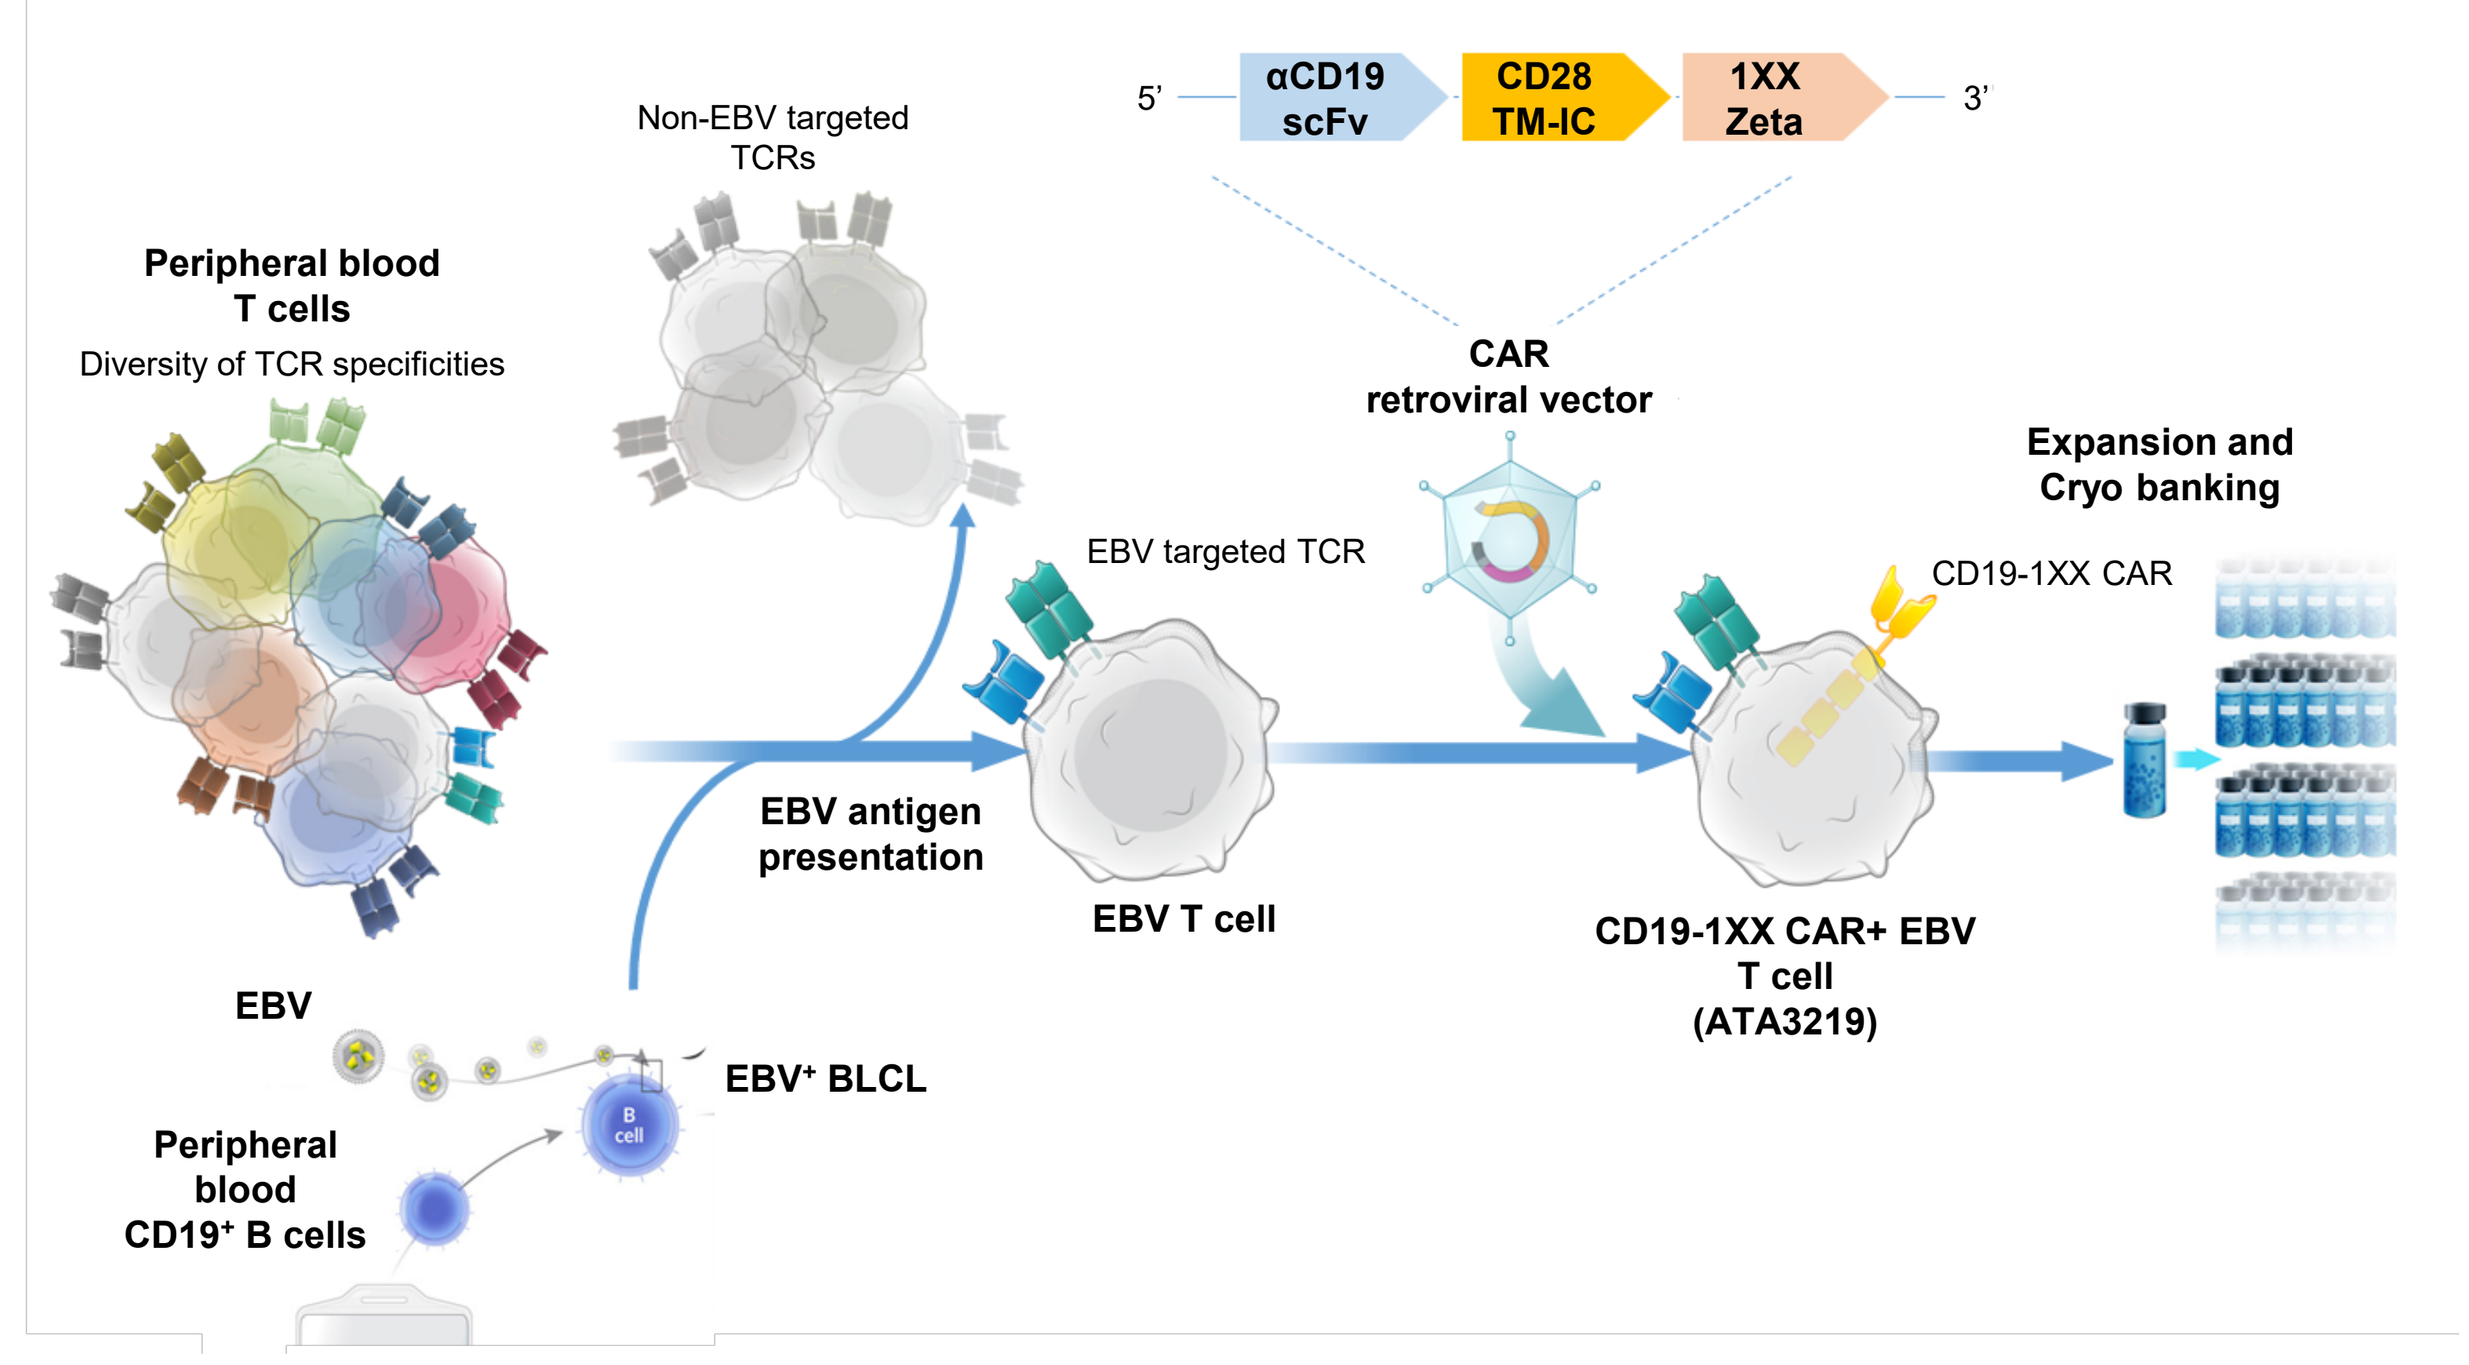
5'
αCD19 scFv
CD28 TM-IC
1XX Zeta
3'
Non-EBV targeted TCRs
Peripheral blood T cells
Diversity of TCR specificities
CAR retroviral vector
EBV targeted TCR
Expansion and Cryo banking
CD19-1XX CAR
EBV antigen presentation
EBV T cell
CD19-1XX CAR+ EBV T cell (ATA3219)
EBV
EBV+ BLCL
Peripheral blood CD19+ B cells

RESULTS

ATA3219 demonstrates stable CAR expression and maintains robust central memory phenotype

Figure 2. Flow cytometric analyses of ATA3219 CAR expression from 4 different donors demonstrated the successful and stable transduction of EBV T cells with ATA3219 retroviral vector (A). Vector copy number analyses showed consistently fewer than 5 vector copies per genome. CD4 and CD8 distribution showed ATA3219 to be predominantly CD8+ (B). Analyses of memory markers CD45RO and CCR7 showed ATA3219 to maintain a robust central memory population compared with autologous benchmark CAR T cells (C). Bars represent standard error of the mean. The gating strategy on live CD3+ cells for the auto benchmark and ATA3219 product from a representative donor is shown in (D).

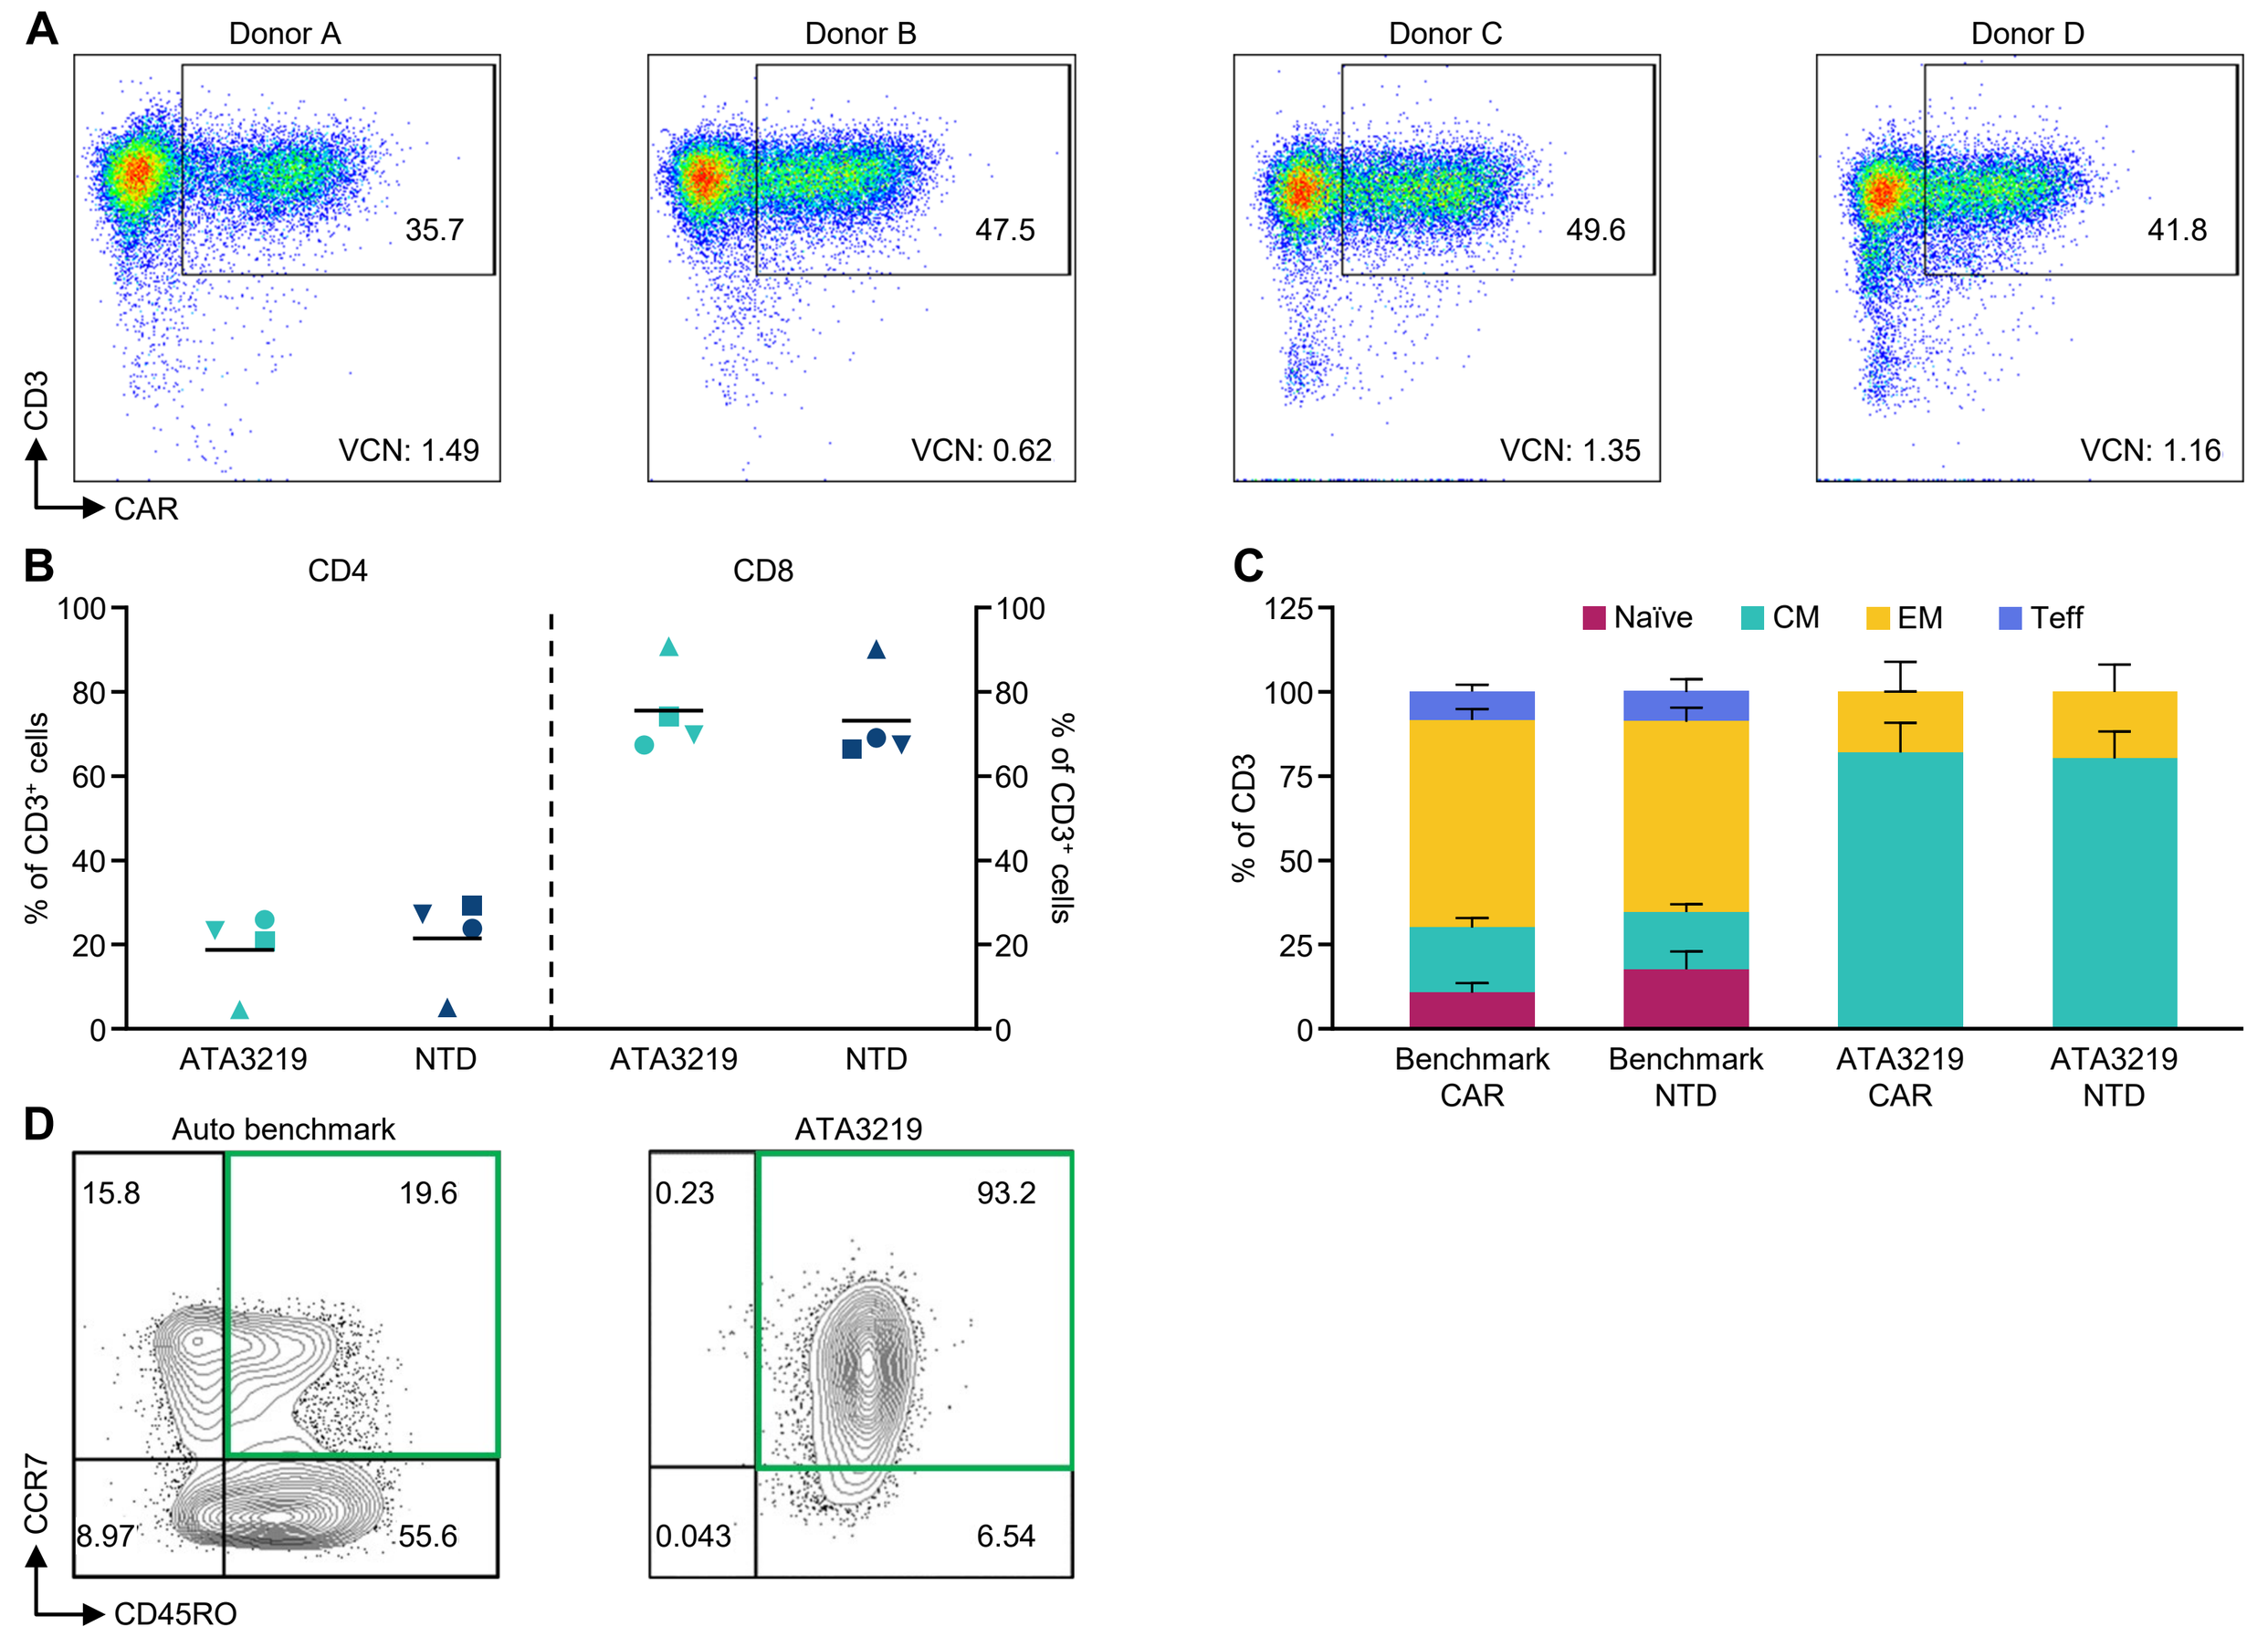
A
Donor A
Donor B
Donor C
Donor D
35.7
47.5
49.6
41.8
VCN: 1.49
VCN: 0.62
VCN: 1.35
VCN: 1.16
CD3
CAR
B
CD4
CD8
% of CD3+ cells
ATA3219
NTD
C
Naïve
CM
EM
Teff
% of CD3
Benchmark CAR
Benchmark NTD
ATA3219 CAR
ATA3219 NTD
D
Auto benchmark
15.8
19.6
8.97
55.6
0.23
93.2
0.043
6.54
CCR7
CD45RO

ATA3219 demonstrates uniform specific functional activity against CD19+ target lines

Figure 3. Specific functional activity against CD19-expressing target lines was measured after a 24- or 48-hour co-culture at the indicated E:T ratios using Nalm-6 (A), CD19-K562 (B) and WT-K562 (C) targets. ATA3219 generated from multiple donors exhibited uniform antigen-specific cytolytic activity against CD19+ target cell lines in a dose-dependent manner compared with NTD T cells. Enumeration of Nalm-6 tumor targets and CAR T cells following repeated serial stimulation (denoted by vertical dotted lines) demonstrated comparable tumor killing (D) and functional persistence (E) compared with autologous benchmark CAR T cells.

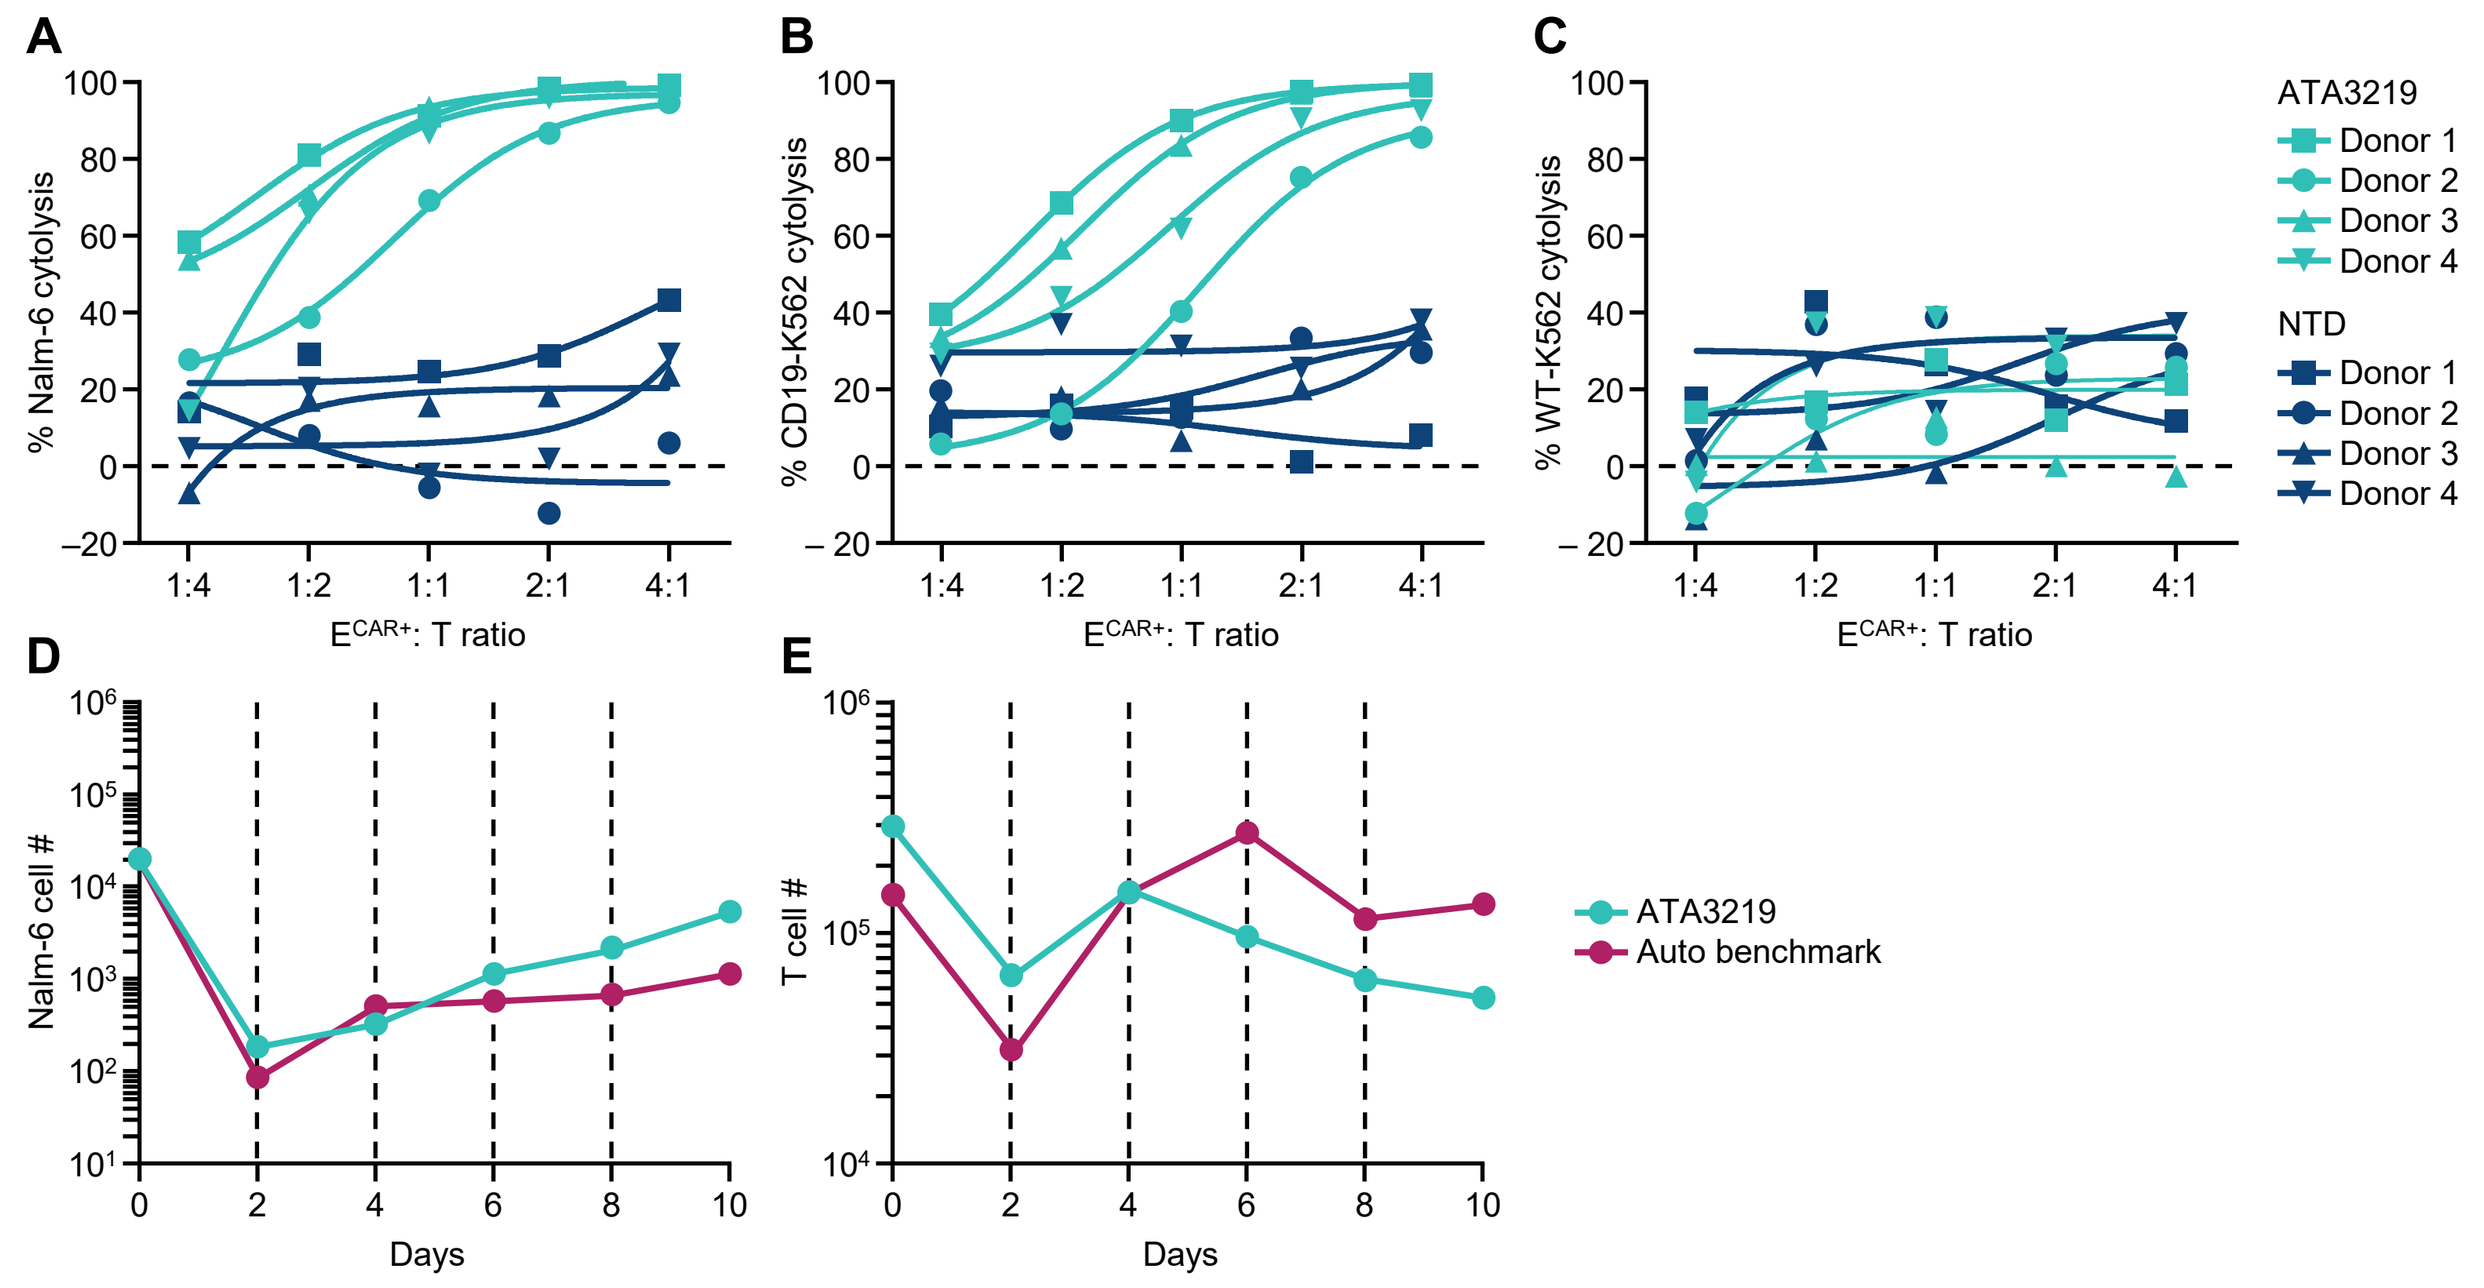
% Nalm-6 cytolysis
% CD19-K562 cytolysis
% WT-K562 cytolysis
1:4
1:2
1:1
2:1
4:1
ECAR+: T ratio
ATA3219
Donor 1
Donor 2
Donor 3
Donor 4
NTD
Donor 1
Donor 2
Donor 3
Donor 4
Nalm-6 cell #
T cell #
Days
ATA3219
Auto benchmark

NTD = non-transduced

ATA3219 demonstrates antigen-specific cytokine secretion and proliferation

Figure 4. ATA3219 demonstrated inflammatory and stimulatory cytokine secretion with higher levels of IFN-γ (A), TNF-α (B) and macrophage inflammatory protein (MIP-1α) (C) in response to CD19+ target lines compared with NTD EBV T cells. A higher percentage of proliferating ATA3219 cells was measured against CD19+ targets compared to fewer dividing NTD EBV T cells (D).

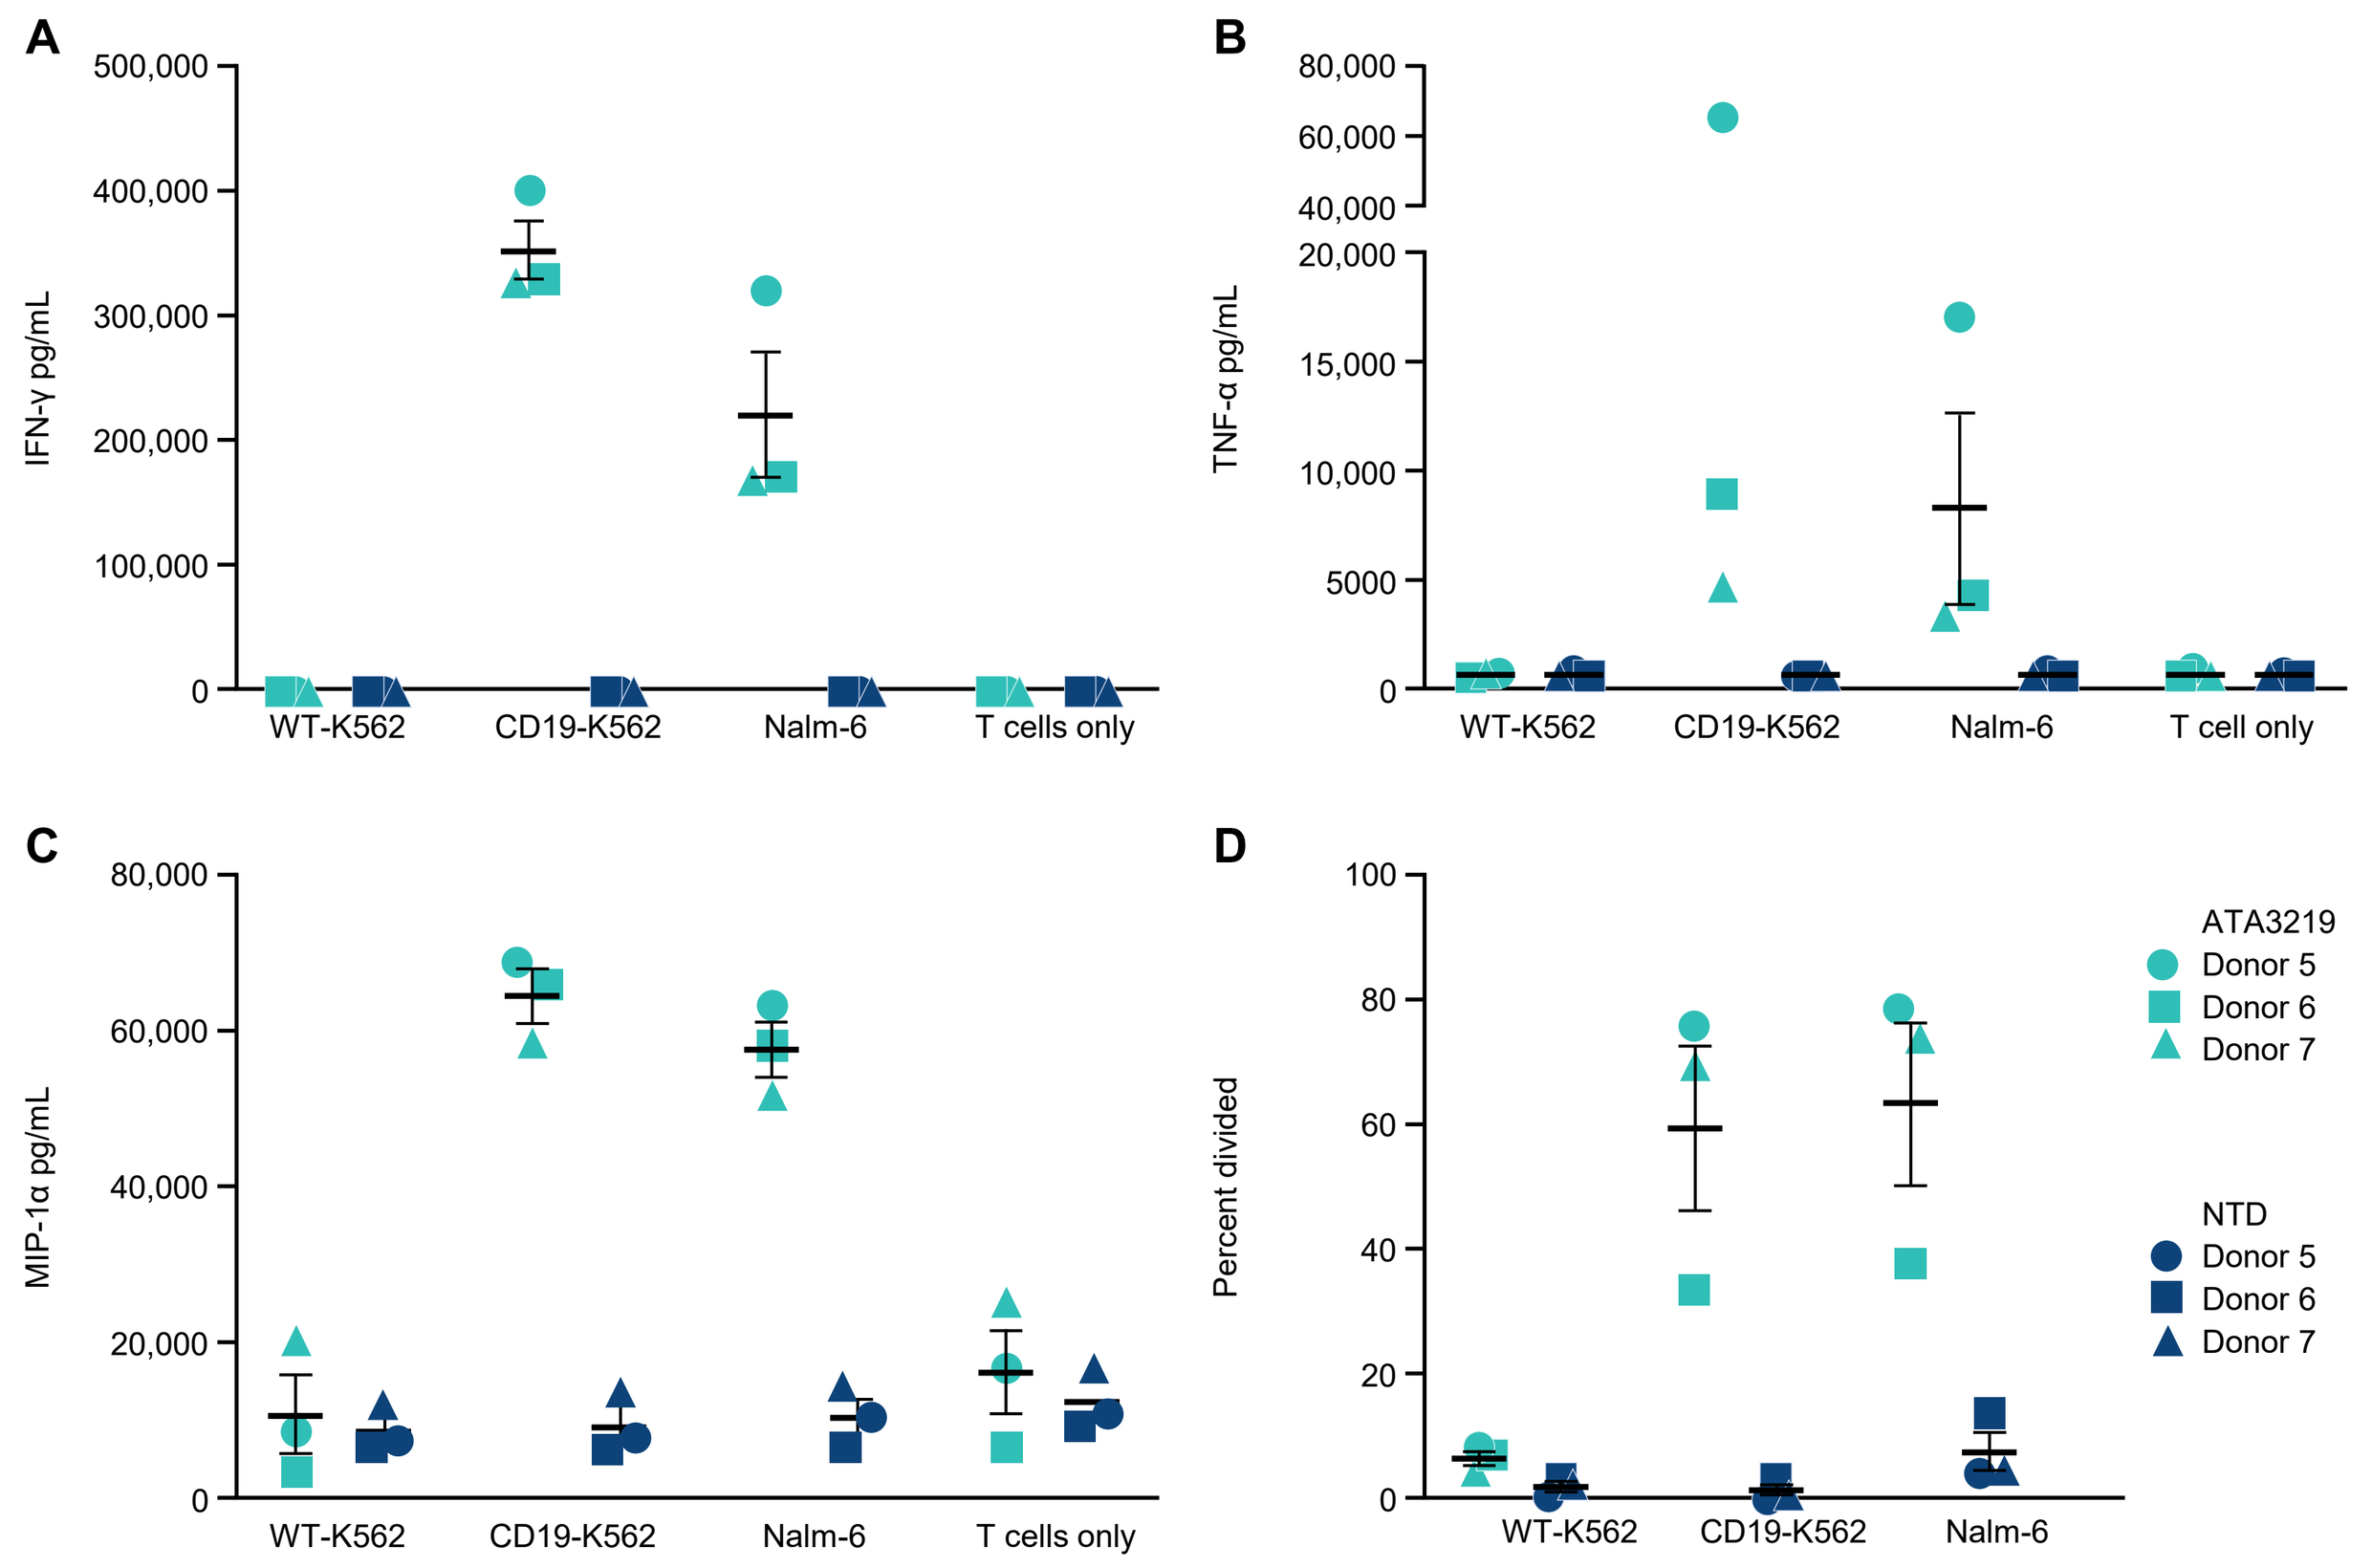
IFN-γ pg/mL
TNF-α pg/mL
MIP-1α pg/mL
Percent divided
WT-K562
CD19-K562
Nalm-6
T cells only
ATA3219
Donor 5
Donor 6
Donor 7
NTD
Donor 5
Donor 6
Donor 7

ATA3219 demonstrates minimal alloreactivity against HLA-mismatched targets

Figure 5. Alloreactive potential was measured through the release of 51Cr from labeled HLA-matched (auto PHAb) and mismatched PHA blasts (cMM PHAb) after 4 hours in co-culture with ATA3219. Minimal alloreactive cytolysis was observed compared to CD19-K562 targets (A). The maintenance of ATA3219 EBV specificity was evaluated using the xCELLigence Real Time Cell Analysis (RTCA). Autologous EBV+ BLCLs were lysed by both ATA3219 and NTD EBV T cells via CD19 and/or HLA-restricted EBV recognition (B), but only complete HLA-mismatched (cMM) EBV+ BLCLs were lysed by ATA3219 via HLA-independent CD19 CAR-directed lysis (C and D).

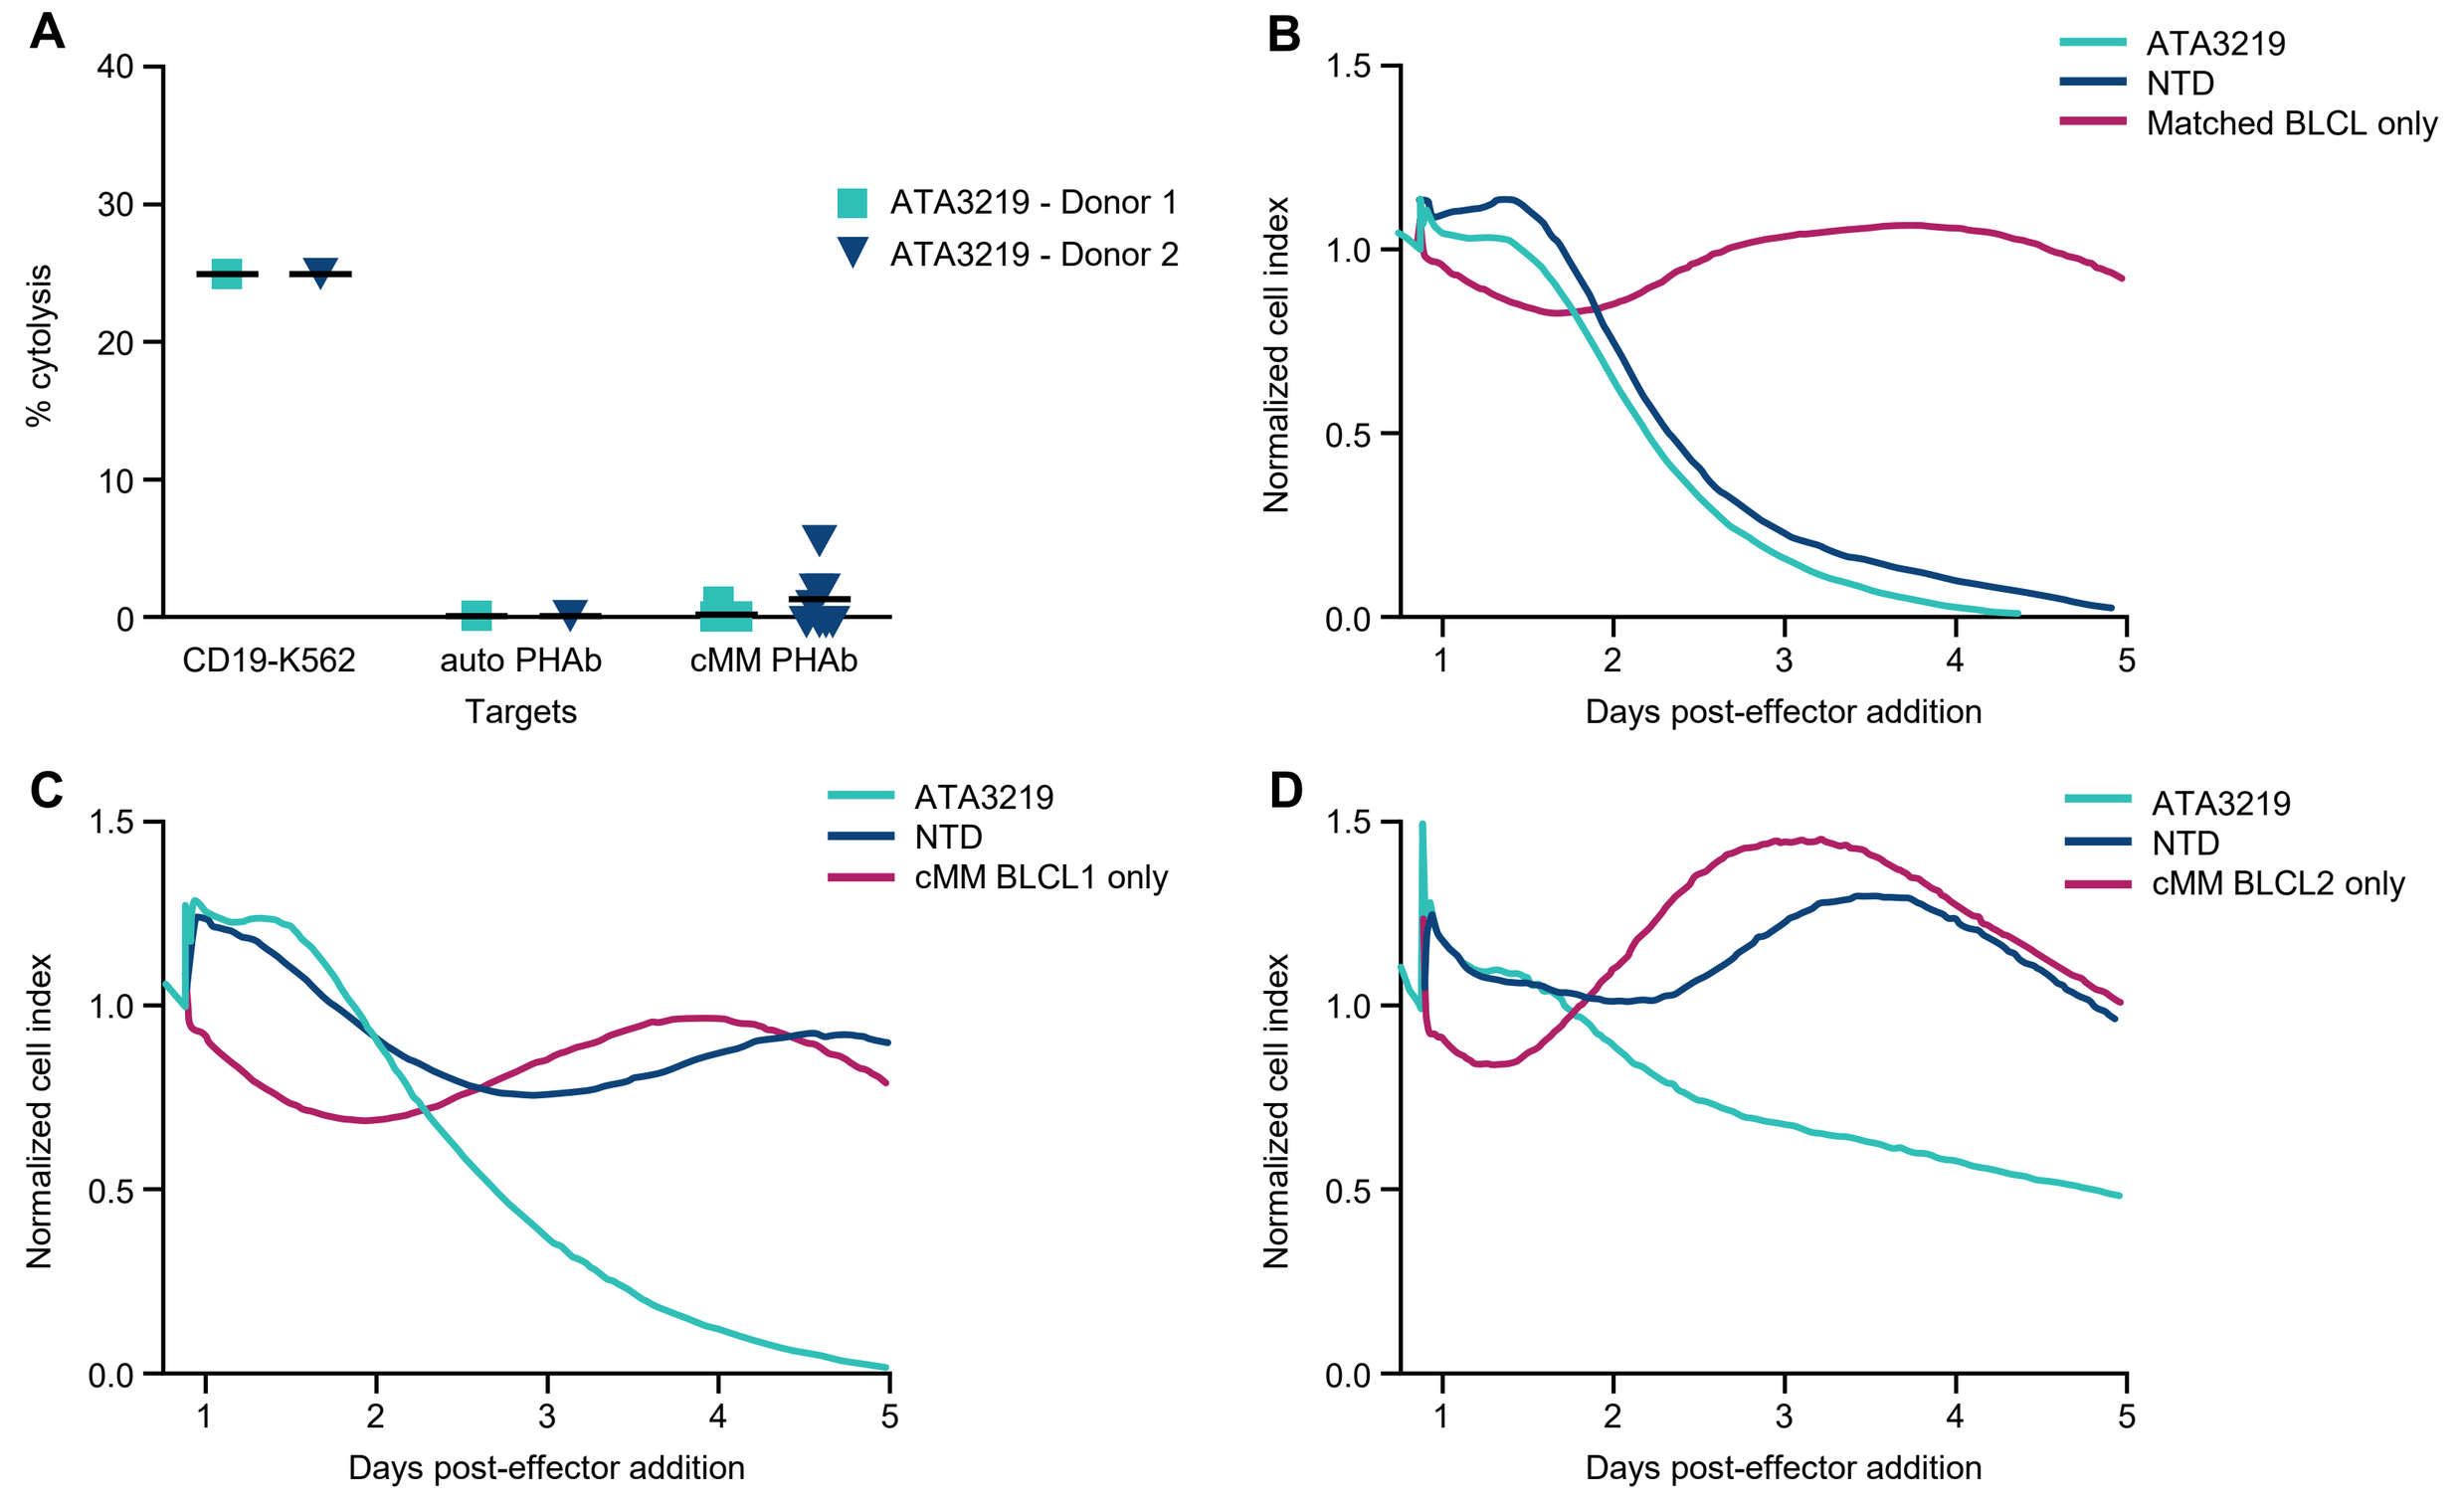
% cytolysis
ATA3219 - Donor 1
ATA3219 - Donor 2
CD19-K562
auto PHAb
cMM PHAb
Targets
Normalized cell index
Days post-effector addition
ATA3219
NTD
Matched BLCL only
cMM BLCL1 only
cMM BLCL2 only

PHA = phytohemagglutinin

Single and multiple dosing of ATA3219 demonstrates potent tumor rejection and corresponds with robust *in vivo* expansion without exogenous cytokine support

Figure 6. NOD scid gamma (NSG) mice were intravenously implanted with 0.5x10[6] Nalm-6 cells on day 0. Mice were randomized into different groups by bioluminescence imaging (BLI) and treated with a single (top row) or multiple (bottom row) IV injections of freshly thawed ATA3219 or NTD EBV T cells, as indicated by the vertical dotted lines. BLI radiance for each mouse (n=8) was measured post tumor implant (A). The absolute count of circulating human T cells (B) and vector copy number (C) were measured. No treatment-related toxicities were observed following single or multiple dosing. Horizontal dotted line represents the VCN of ATA3219 at the time of infusion.

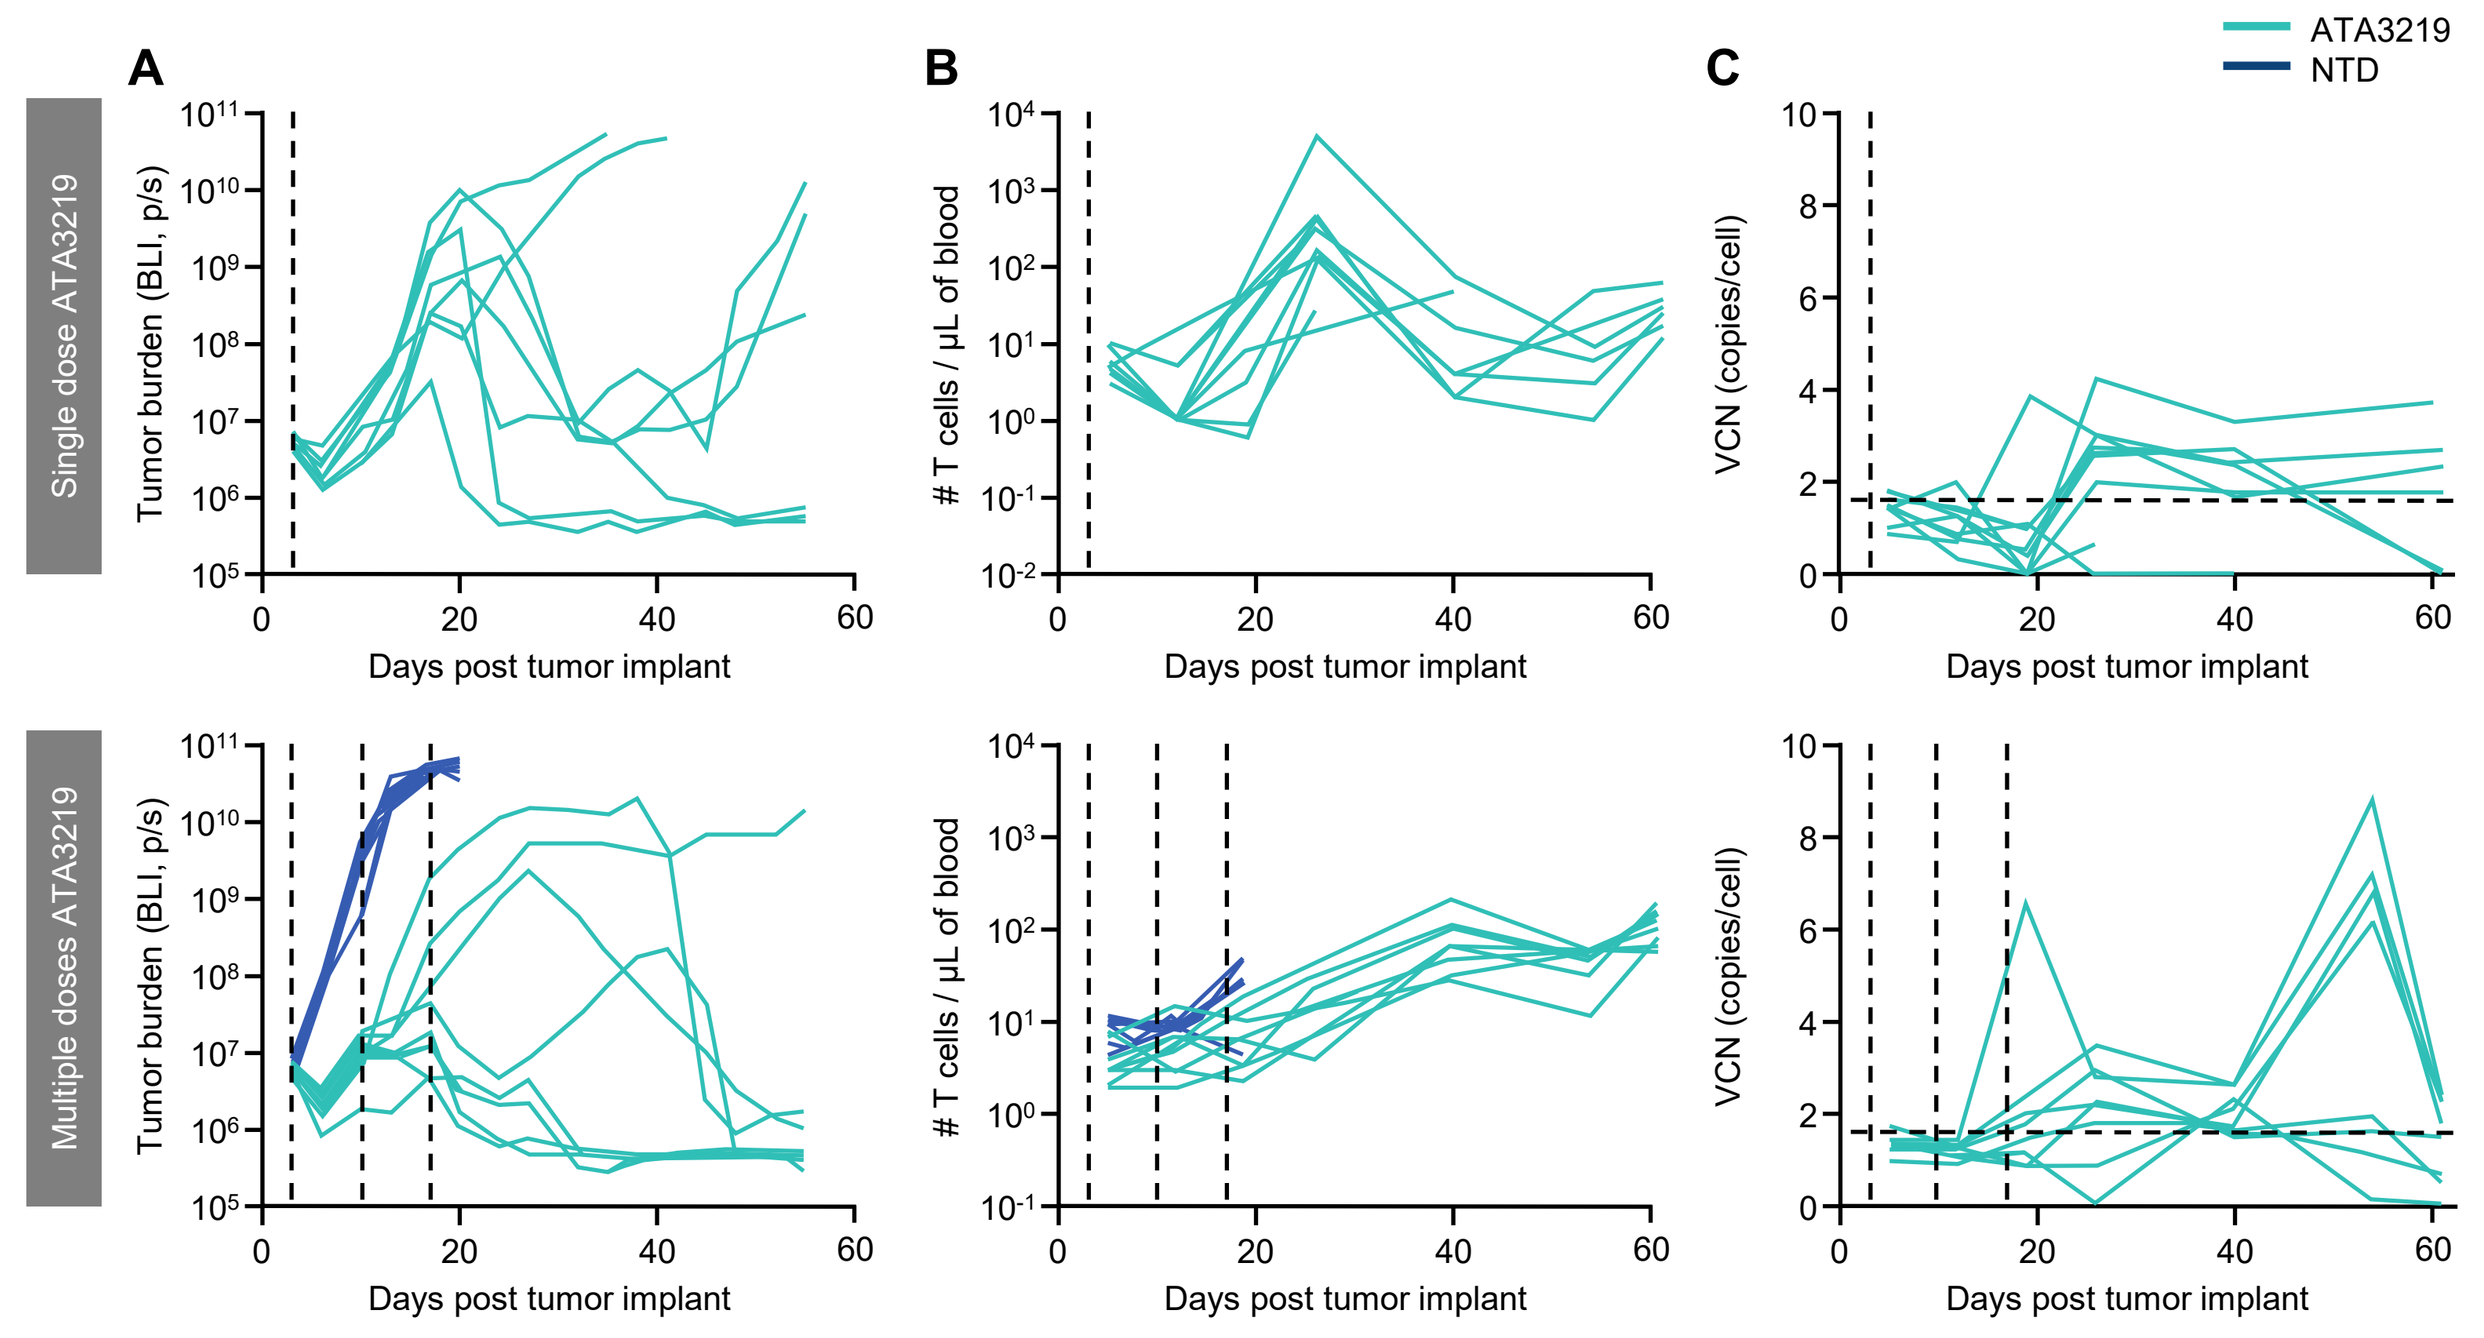
Single dose ATA3219
Multiple doses ATA3219
Tumor burden (BLI, p/s)
T cells / µL of blood
VCN (copies/cell)
Days post tumor implant
ATA3219
NTD

ATA3219 demonstrates superior *in vivo* anti-tumor activity and functional persistence compared to auto benchmark CAR T cells

Figure 7. NSG mice were intravenously implanted with 0.5x10[6] Nalm-6 cells on day 0. Mice were randomized into different groups by BLI and treated with a single intravenous injection of freshly thawed T cells. BLI radiance for each mouse (n=5-8) was measured post-tumor implantation (A). Group median BLI (B), survival (C) and vector copy number, normalized to the input number of CAR copies/cell, (D) of the groups treated with ATA3219 or auto benchmark CAR T cells were analyzed. Animals treated with benchmark CAR T cells showed alloreactive expansion of non-CAR T cells after 40 days.

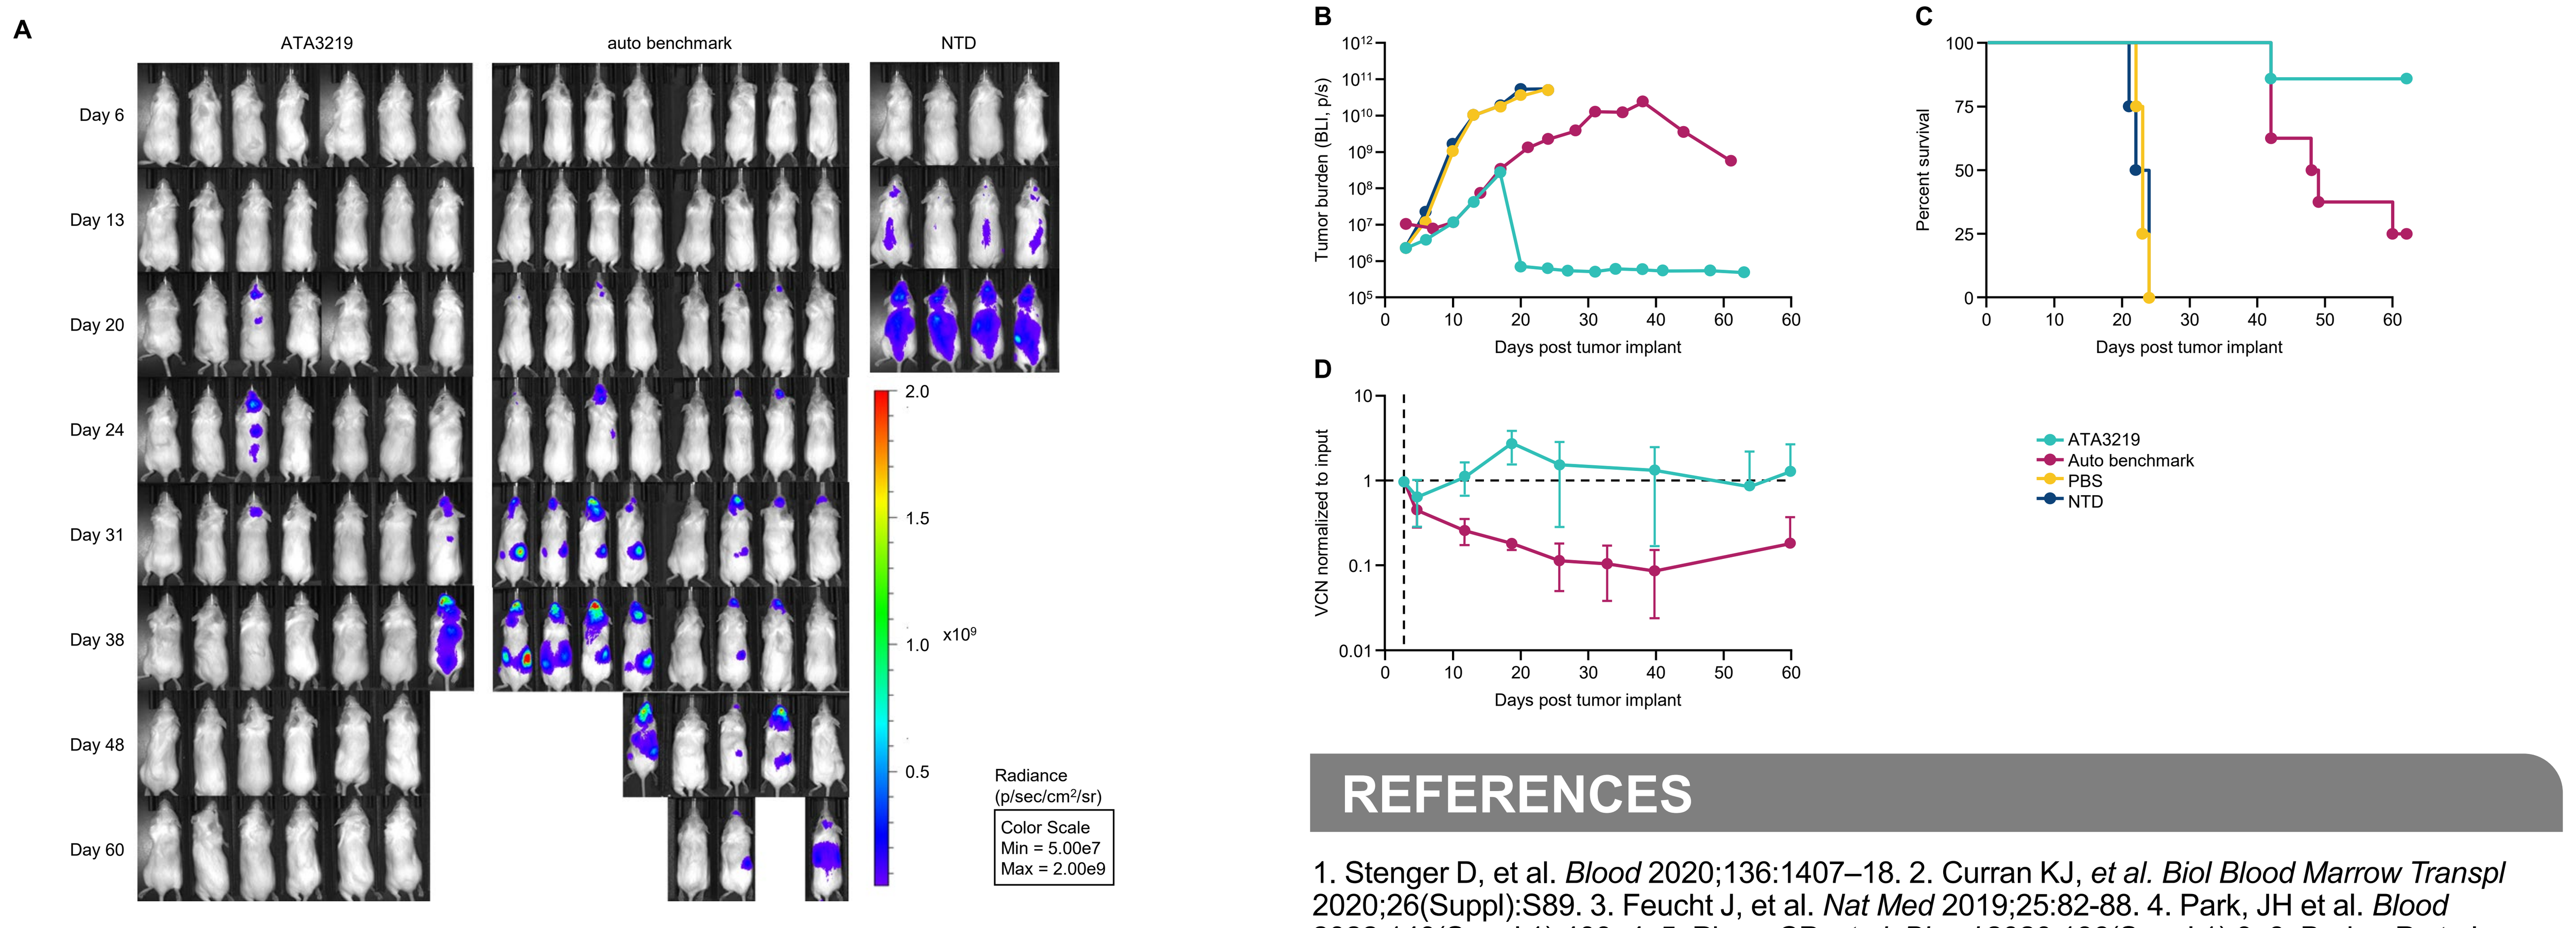
ATA3219
auto benchmark
NTD
Day 6
Day 13
Day 20
Day 24
Day 31
Day 38
Day 48
Day 60
Radiance (p/sec/cm2/sr)
Color Scale
Min = 5.00e7
Max = 2.00e9
x10⁹
Tumor burden (BLI, p/s)
Percent survival
VCN normalized to input
Days post tumor implant
ATA3219
Auto benchmark
PBS
NTD

CONCLUSIONS

- **ATA3219 is an off-the-shelf allogeneic T-cell therapy targeting CD19 via next-generation CAR (1XX) built on Atara's EBV T-cell platform.**
- **We have engineered allogeneic EBV T cells to stably express CD19 CAR with a 1XX costimulatory domain with high efficiency (Fig 2).**
- **Optimization of ATA3219 enriches for a predominant central memory phenotype (Fig 2).**
- **ATA3219 demonstrates durable CD19 antigen-specific cytotoxic activity (Fig 3), polyfunctional activation profile and proliferation potential (Fig 4).**
- **ATA3219 demonstrates minimal alloreactivity against HLA mismatched targets and shows HLA-independent activity against CD19+ targets (Fig 5).**
- **Single and multiple dosing of ATA3219 is well tolerated and demonstrates potent tumor rejection corresponding with *in vivo* expansion in the absence of exogenous cytokine support (Fig 6).**
- **ATA3219 shows superior robust anti-tumor activity, superior survival and persistence compared to autologous benchmark CAR T cells with no observed toxicity or alloreactivity (Fig 7).**
- **In summary, this updated preclinical dataset demonstrates the flexibility of our EBV T-cell generation process through improvements in the polyfunctional phenotype, potent expansion and targeting against B cell tumors with low alloreactivity. These findings support advancing ATA3219 to clinical evaluation.**

REFERENCES

1. Stenger D, et al. *Blood* 2020;136:1407–18. 2. Curran KJ, *et al. Biol Blood Marrow Transpl* 2020;26(Suppl):S89. 3. Feucht J, et al. *Nat Med* 2019;25:82-88. 4. Park, JH et al. *Blood* 2022;140(Suppl 1):403–4. 5. Pham CP, *et al. Blood* 2020;136(Suppl 1):9. 6. Barba, P et al. *Blood* 2022;140(Suppl 1):1056–9.

Presented at the Transplantation & Cellular Therapy Meetings of ASTCT and CIBMTR (2023), February 15-19, Orlando, FL